FibroTest™ Is an Independent Predictor of Early and Sustained Virologic Response in Nonresponder Patients With Chronic Hepatitis C: Results From the EPIC³ Study

T. Poynard,[1] M. Munteanu,[2] M. Colombo,[3] J. Bruix,[4] E. Schiff,[5] R. Terg,[6] S. Flamm,[7] R. Moreno-Otero,[8] F. Carrilho,[9] W. Schmidt,[10] T. Berg,[11] T. McGarrity,[12] E. J. Heathcote,[13] F. Gonçales,[14] M. Diago,[15] A. Craxi,[16] M. Silva,[17] P. Bedossa,[18] P. Mukhopadhyay,[19] L. Griffel,[19] M. Burroughs,[19] C. Brass,[19] J. K. Albrecht[19] for the EPIC³ Study Group

[1]APHP-UPMC Liver Center, Paris, France; [2]BioPredictive, Paris, France; [3]1st Division of Gastroenterology, Fondazione IRCCS Maggiore Hospital, University of Milan, Milan, Italy; [4]Centro de Investigación Biomédica en Red de Enfermedades Hepaticas y Digestivas, Liver Unit, Hospital Clinic, University of Barcelona, Barcelona, Spain; [5]University of Miami School of Medicine, Miami, Florida, USA; [6]Hospital Municipal de Gastroenterologia Dr. Bonorino Udaondo, Capital Federal, Argentina; [7]Northwestern University, Chicago, Illinois, USA; [8]Hospital Universitario de la Princesa and Centro de Investigación Biomédica en Red de Enfermedades Hepaticas y Digestivas (Instituto de Salud Carlos III), Madrid, Spain; [9]Department of Gastroenterology, University of São Paulo School of Medicine, São Paulo, Brazil; [10]University of Iowa Hospitals and Clinics, Iowa City, Iowa, USA; [11]Charité, Campus Virchow Klinikum, Universitätsmedizin Berlin, Berlin, Germany; [12]Milton S. Hershey Medical Center, Hershey, Pennsylvania, USA; [13]University Health Network, Toronto, ON, Canada; [14]Department of Medical Clinical, Faculty of Medical Sciences, University of Campinas, Campinas, Brazil; [15]Hospital General Universitario de Valencia, Valencia, Spain; [16]GI and Liver Unit, DIBIMIS, University of Palermo, Palermo, Italy; [17]Hospital Universitario Austral, Pilar, Argentina; [18]Anatomie Pathologique, Hôpital Beaujon, Clichy, France; and [19]Schering-Plough Research Institute, Kenilworth, New Jersey, USA

Abstract

Background: EPIC³, a prospective, international, multicenter, open-label study, demonstrated the efficacy and safety of PEG-2b plus weight-based ribavirin in chronic hepatitis C patients with significant fibrosis who previously failed any interferon-alfa/ribavirin combination therapy. The aim of the present study was to assess if FibroTest™, a validated noninvasive marker of fibrosis in naive patients, is also an alternative to biopsy as a predictor of early virologic (EVR) and sustained virologic response (SVR) in previously treated patients.

Methods: Patients enrolled in EPIC³ were included in this evaluation if they had available 12 weeks' virology (TW12), interpretable baseline FT, and biopsy. Univariate (UV) and multivariate (MV) analysis for SVR was performed using a complete model that included FT and biopsy.

Results: Of 2312 patients enrolled, 1459 had available baseline FT, biopsy, and complete data. Baseline characteristics: 70% male, median age 51 yr; METAVIR, 28% F2, 29% F3, and 43% F4; previous relapsers, 29%; previous PEG regimen 41%; and high (>6 × 10^5 IU/mL) baseline viral load (BVL), 64%. Patients received PEG-2b 1.5 µg/kg/wk plus weight-based ribavirin (800-1400 mg/d) for 12-18 weeks; 506 (35%) had undetectable serum hepatitis C virus (HCV)-RNA at TW12 (TW12 neg); 678 (46%) were treated 48 weeks, with 24 weeks follow-up. SVR was 58% among 506 patients TW12 neg, 13% among 108 patients with detectable but ≥2 log drop in viral load and 0% among 43 patients with <2 log drop. The accuracy of FT for the diagnosis of fibrosis was similar to previous validations: area under the ROC curve for the diagnosis of F4 vs F2=0.80 (P<.00001), when standardized according to fibrosis stages' prevalences. Five baseline factors were significantly (P<.001) associated with SVR, in UV and MV analyses (odds ratio: UV/MV): fibrosis stage estimated using FT (4.5/5.9) or biopsy (1.5/1.6), genotype 2/3 (4.5/5.1), BVL (1.5/1.3), prior relapse (1.6/1.6), and previous treatment with non-PEG (2.6/2.0). Similarly, these same factors were significantly (P≤.001) associated with EVR: fibrosis stage estimated using FT (3.8/4.8) or biopsy (1.2/1.3), genotype 2/3 (11/9), BVL (1.6/1.3), prior relapse (6.2/6.5), and previous treatment with non-PEG (2.6/2.0). Among patients TW12 neg (n=506), only two factors remained highly predictive of SVR by MV analysis (P≤.001): genotype 2/3 (odds ratio=2.9), and fibrosis estimated with FT (4.3) or by biopsy (1.5).

Conclusions: FibroTest™ is a noninvasive alternative to biopsy for the prediction of EVR at 12 weeks and for prediction of SVR, in patients with previous failures, retreated with peginterferon alfa-2b and ribavirin, both at baseline and at week 12.

Background

- Assessment of fibrosis stage is useful for predicting therapeutic outcomes in patients undergoing treatment for chronic hepatitis C.
 - Because of the limitations of liver biopsy, several noninvasive methods have been developed as alternatives.
- FibroTest™ (FT; BioPredictive, Paris, France) is a set of noninvasive biomarkers of liver fibrosis that has been extensively validated in patients with chronic hepatitis C.[1,2]
 - Diagnostic and prognostic values of FT are similar to those of a biopsy specimen 25 mm in length.[3]
 - FT has been approved in France for first-line assessment of fibrosis and cirrhosis in patients with chronic hepatitis C.[4]
- FT has been extensively studied during treatment of treatment-naive patients[3,5-8]; however, it has not been assessed during retreatment of previous nonresponders or relapsers to standard treatment.

Aim

- To validate the utility of FT as an alternative to biopsy in predicting sustained virologic response (SVR) in nonresponders in Evaluation of PegIntron in Control of Hepatitis C Cirrhosis (EPIC³), a large prospective clinical trial.

Patients and Methods

- EPIC³ was a multiphase clinical program designed to evaluate the retreatment of patients with chronic hepatitis C who had significant fibrosis/cirrhosis and in whom previous treatment with nonpegylated or pegylated interferon (PEG-IFN) alfa plus ribavirin was ineffective.

Patients

- Patients aged 18 to 65 years who had chronic hepatitis C and in whom combination therapy with nonpegylated or PEG-IFN alfa plus ribavirin therapy failed were eligible to enroll in EPIC³.
- Additional inclusion criteria included hepatic fibrosis documented by historical biopsy showing at least portal fibrosis with few septa (METAVIR score ≥F2).
- Patients enrolled in EPIC³ were included in the present FT evaluation if they had available treatment week 12 (TW12) virology data, interpretable baseline FT results, and biopsy results (diagnostic population).

Study Design

- EPIC³ was an international, prospective, open-label clinical trial conducted at 133 sites.
- Patients received PEG-IFN alfa-2b 1.5 µg/kg/wk and daily weight-based ribavirin (800-1400 mg/day) for up to 48 weeks.
- Patients with detectable HCV-RNA at TW12 were offered randomization into a maintenance treatment phase.
- The study was approved by the ethics committee at each participating institution and was conducted in accordance with good clinical practice and the tenets of the Declaration of Helsinki.

Assessments

- FibroTest™ was measured at screening.
 - Samples were blindly assessed without knowledge of any patient characteristics.
 - FT combined the following five markers: α2-macroglobulin, haptoglobin, γ-glutamyl transpeptidase, total bilirubin, and apolipoprotein A1.
- Pretreatment liver biopsy specimens were scored by a single pathologist (P.B.) using METAVIR criteria.
- Plasma HCV-RNA was measured at screening, at weeks 12, 24, and 48 of treatment, and at 12 and 24 weeks after treatment.
 - The primary efficacy end point of the therapeutic study was SVR, defined as undetectable serum HCV-RNA 24 weeks after treatment.
 - A secondary efficacy end point was the difference in SVR rates by fibrosis score.

Analyses

- Diagnostic values of FT were assessed by the receiver-operating characteristic (ROC) curves.
 - Respective overall diagnostic values were compared using the area under the ROC curve (AUROC).
 - AUROC values were compared with 0.50 (no diagnostic value) and with AUROCs observed in an integrated database of treatment-naive patients with chronic hepatitis C.[9]
- Estimates of AUROCs and comparisons between AUROCs used empirical nonparametric methods.
 - It has been demonstrated that the FT AUROC varied from 0.67 to 0.98 because of the different spectrum of stages included in advanced fibrosis and in nonadvanced fibrosis.[10] To prevent the risk of spectrum bias from different prevalences of fibrosis stage between studies, we used the previously validated method of standardization.[10]
 - The previously validated adjusted FT AUROC was used as the gold standard to prevent the errors inherent when considering a biopsy specimen of small length.[11]
- Univariate and multivariate analyses for SVR were performed with the use of a complete model that included FT and biopsy.
 - Logistic regression analyses were performed to assess the effect of the prognostic factors on SVR rates.
 - SVR rates were also summarized by baseline METAVIR fibrosis score (F2, F3, and F4), estimated by FT and biopsy.
 - The Armitage S trend test was used to compare the proportion of patients who attained SVR.

Results

Patient Characteristics

- In total, 2312 patients were enrolled in the EPIC³ study. Of those, 1459 patients (diagnostic population) had available baseline FT data, biopsy results, and complete virology data and were included in the present evaluation. Four patients were excluded because FT results were not interpretable (Figure 1).

Figure 1. Patient flow diagram. [a] Continued in protocol at the discretion of the investigator. [b] The decrease in HCV-RNA at TW12 was not quantifiable for 21 subjects; 4/21 (19%) attained SVR. SVR = sustained virologic response; TW12 = treatment week 12.

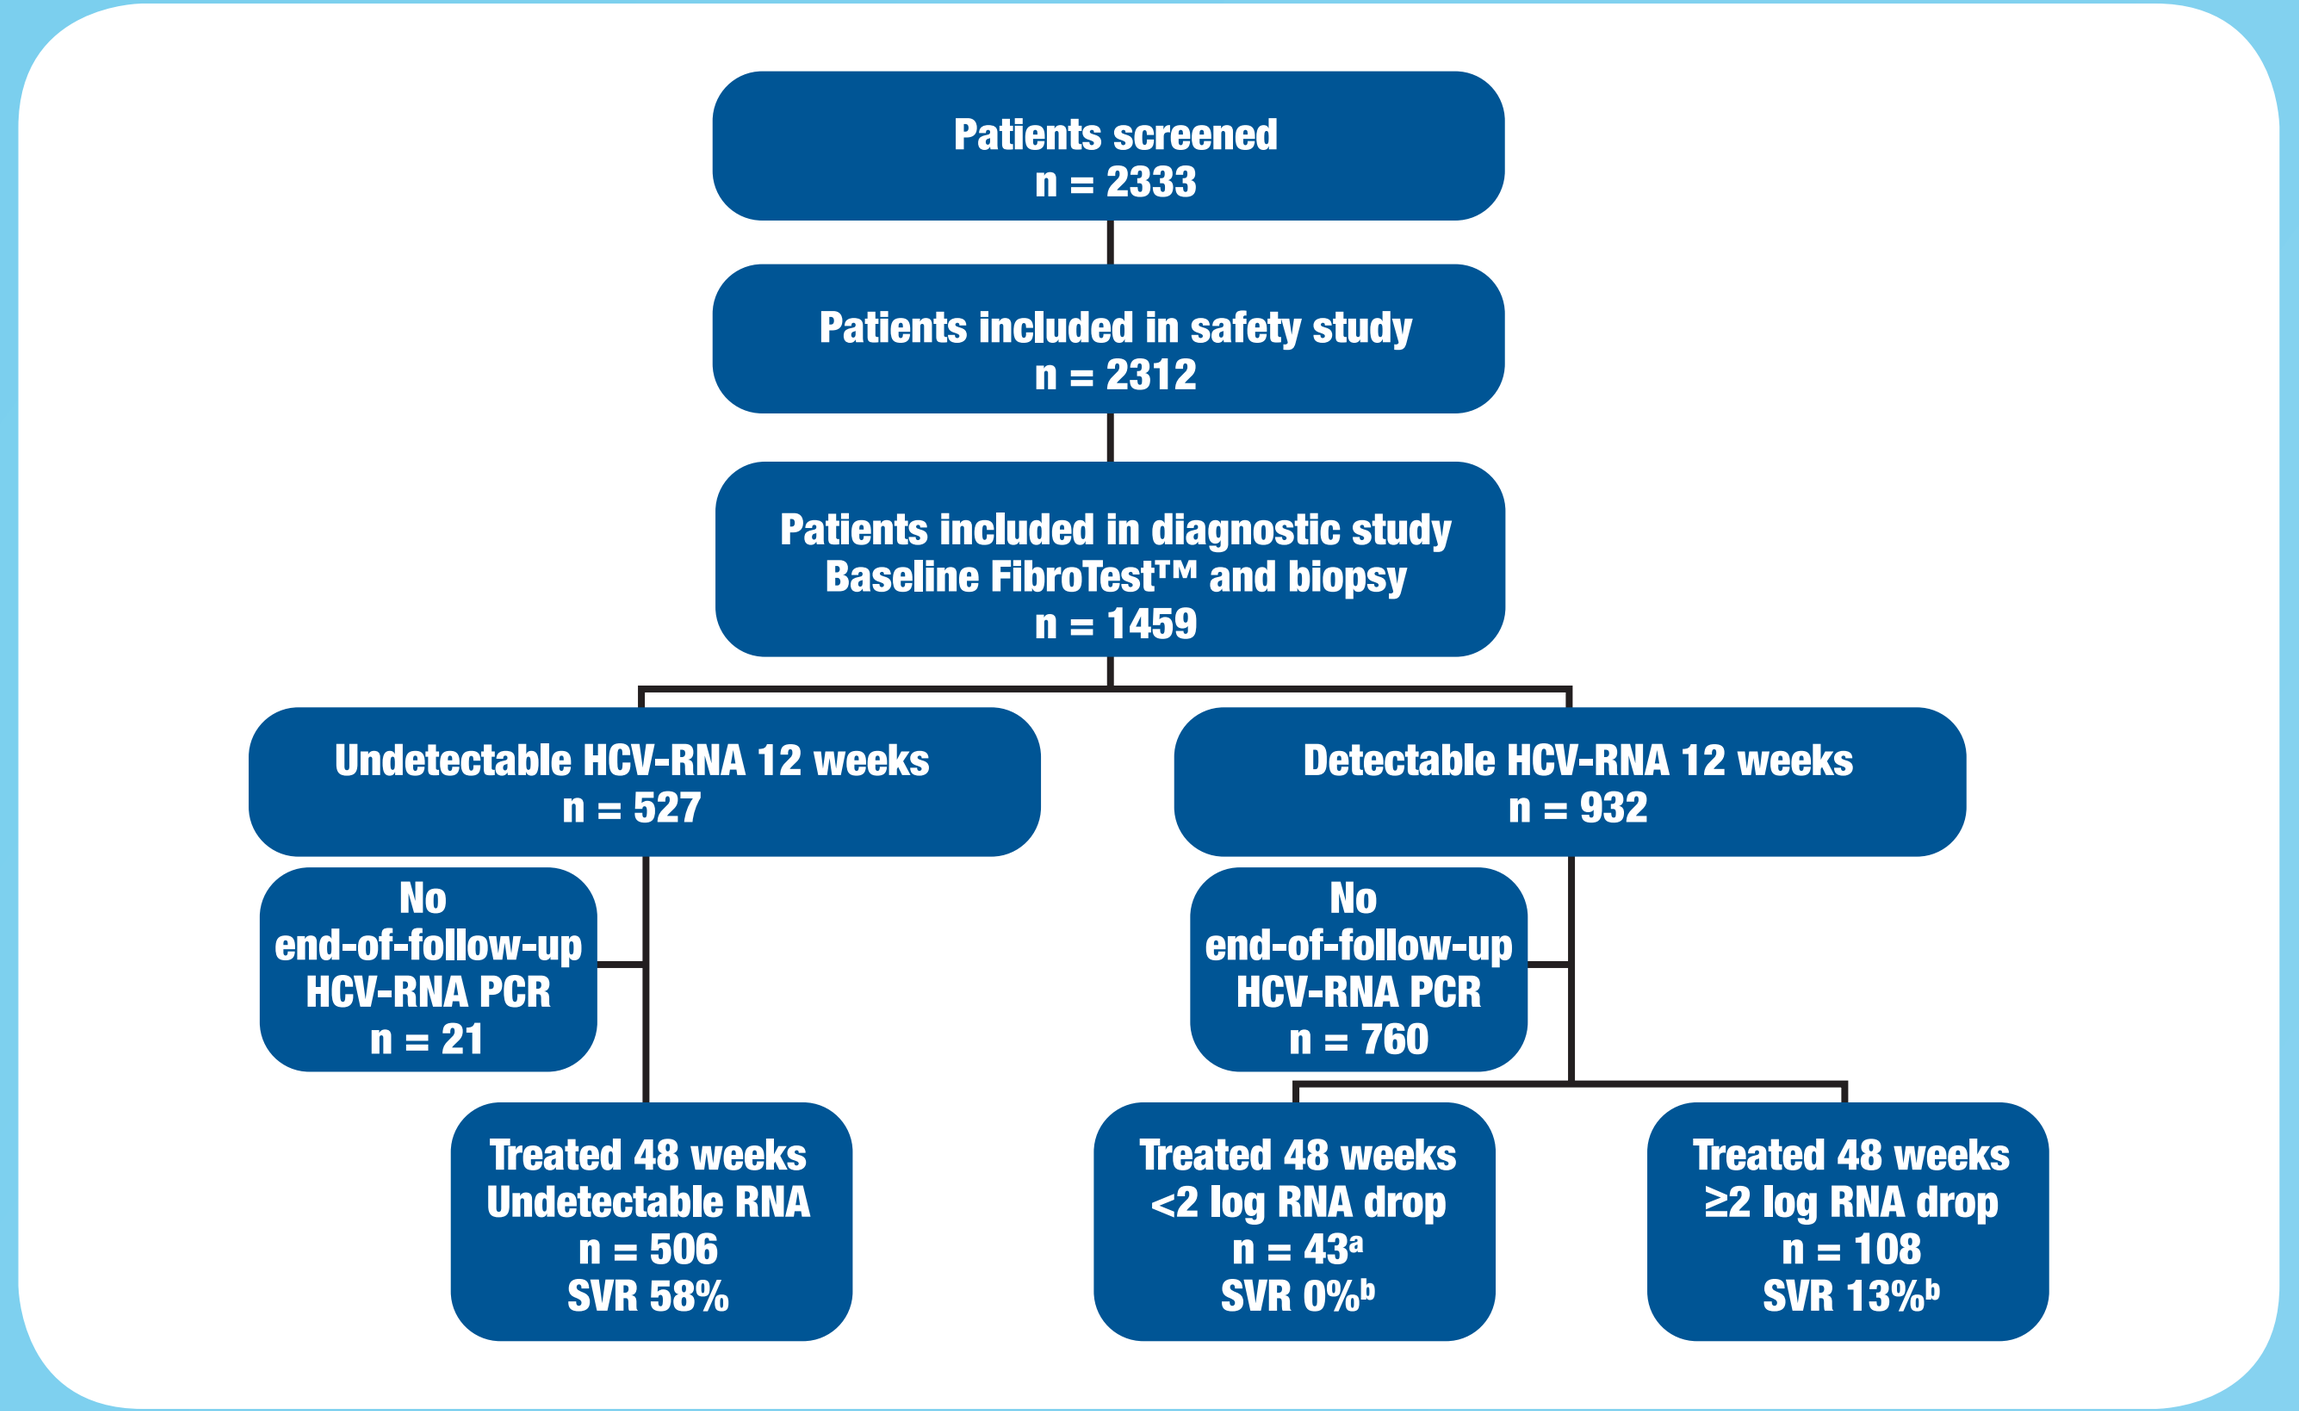
Patients screened
n = 2333
Patients included in safety study
n = 2312
Patients included in diagnostic study
Baseline FibroTest™ and biopsy
n = 1459
Undetectable HCV-RNA 12 weeks
n = 527
Detectable HCV-RNA 12 weeks
n = 932
No
end-of-follow-up
HCV-RNA PCR
n = 21
No
end-of-follow-up
HCV-RNA PCR
n = 760
Treated 48 weeks
Undetectable RNA
n = 506
SVR 58%
Treated 48 weeks
<2 log RNA drop
n = 43a
SVR 0%b
Treated 48 weeks
≥2 log RNA drop
n = 108
SVR 13%b

- There was no significant difference in baseline characteristics between the safety population (all patients enrolled in the EPIC³ trial) and the diagnostic population (**Table 1**).

Table 1. Baseline Patient Characteristics of the Safety and Diagnostic Populations

	Safety Population[a] N = 2312	Diagnostic Population n = 1459
Male, n (%)	1650 (71)	1015 (70)
Mean age, y	49.2	50.9
Mean weight, kg	81.1	80.5
Fibrosis stage (METAVIR), n (%)		
Few septa (F2)	658/2308 (29)	410/1459 (28)
Many septa (F3)	676/2308 (29)	424/1459 (29)
Cirrhosis (F4)	974/2308 (42)	625/1459 (43)
Necroinflammatory activity (METAVIR), n (%)		
No activity (A0)	157/2312 (7)	92/1459 (6)
Minimal activity (A1)	1742/2312 (75)	1107/1459 (76)
Moderate activity (A2)	389/2312 (17)	245/1459 (17)
Severe activity (A3)	22/2312 (1)	15/1459 (1)
Genotype, n (%)		
1	1859 (80)	1203 (82)
2	75 (3)	47 (3)
3	293 (13)	168 (12)
4	68 (3)	37 (3)
Missing	17 (1)	4 (<1)
Baseline viral load, n (%)		
≤600,000 IU/mL	853 (37)	524 (36)
>600,000 IU/mL	1451 (63)	935 (64)
Missing	8 (<1)	0 (0)
Previous combination therapy, n (%)		
IFN alfa + ribavirin	1425 (62)	857 (59)
PEG-IFN alfa + ribavirin	865 (37)	602 (41)
No combination therapy	22 (1)	0 (0)
Previous response, n (%)		
Nonresponder	1401 (61)	880 (60)
Relapser	647 (28)	424 (29)
Treatment failure[b]	264 (11)	155 (11)

F = METAVIR score; PEG-IFN = pegylated interferon.
[a] All patients enrolled in the EPIC³ trial.
[b] Per Halfon et al.[2]

Diagnostic Value of FibroTest™

- The accuracy of FT for the diagnosis of fibrosis stage in nonresponders is similar to its accuracy in a previous validation in treatment-naive patients (**Table 2**).[2]

Table 2. Diagnostic Value of FibroTest™ (AUROCs) for Differentiating Between Fibrosis Stages in the Diagnostic Population of the Present Study (Nonresponders) and in Previous Integrated Data (Treatment-Naive Patients)

	FibroTest™ vs Biopsy: Nonresponders	FibroTest™ vs Biopsy: Treatment-Naive[a]
F2 vs F4		
No. patients	1035	376
Observed AUROC (95% CI)	0.75 (0.72-0.78)	0.75 (0.70-0.80)
Standardized AUROC[b]	0.80	0.80
Adjusted on biopsy-length AUROC[c]	0.82	0.82
F2 vs F3		
No. patients	834	364
Observed AUROC (95% CI)	0.65 (0.61-0.68)	0.63 (0.58-0.68)
Standardized AUROC[b]	0.81	0.79
Adjusted on biopsy-length AUROC[c]	0.76	0.73
F3 vs F4		
No. patients	1049	234
Observed AUROC (95% CI)	0.62 (0.59-0.66)	0.65 (0.59-0.71)
Standardized AUROC[b]	0.78	0.81
Adjusted on biopsy-length AUROC[c]	0.72	0.76

AUROC = area under the receiver-operating characteristic curve; CI = confidence interval; F = METAVIR score.
All AUROCs were significant (P < .001) versus random AUROC (0.50). There was no significant difference between the AUROCs of nonresponders and treatment-naive patients.
[a] Per Poynard et al.[9]
[b] Standardized AUROC preventing spectrum bias for eventual comparisons between studies, corresponding to a standard difference of fibrosis stage of 2.5 METAVIR unit.
[c] Gold standard–adjusted AUROC for the given biopsy length preventing the bias of considering biopsy specimen as a gold standard calculated as the ratio between observed AUROC and gold standard (entire liver) AUROC of 16-mm biopsy specimen vs gold standard (0.82 for F3 vs F4 and 0.86 for F2 vs F3).

Virologic Response in the Diagnostic Population

- In total, 506 of 1459 (35%) patients had undetectable serum HCV-RNA at TW12 and had available end-of-follow-up (EOF) HCV-RNA results; 678 (46%) continued participation in the retreatment study and had available EOF HCV-RNA results.
- Overall SVR rate was 21% (312/1459) (**Table 3**).
 - SVR rate was 58% among patient with undetectable HCV-RNA at TW12 (n = 506).
 - SVR rate was 13% among patients with detectable but ≥2 $\log_{10}$ decrease in viral load at week 12 (n = 108).
 - SVR was 0% among patients with <2 $\log_{10}$ decrease (n = 43).
 - These 43 patients were allowed to continue in the retreatment study after week 12 at the discretion of the investigator.
 - The decrease in HCV-RNA at TW12 was not quantifiable for 21 subjects; 4/21 (19%) attained SVR.
- Virologic response rates were similar in the diagnostic population and the overall EPIC³ study population (**Table 3**).

Table 3. Virologic Response Rates in the Diagnostic Population and the Overall EPIC³ Population

	Diagnostic Population (%)	Primary Efficacy Population (%)
cEVR	506/1459 (34.7)	823/2293 (36)
SVR	312/1459 (21)	497/2293 (22)
SVR among patients with cEVR	294/506 (58)	463/823 (56)
SVR among patients with pEVR	14/108 (13)	23/188 (12)

cEVR = complete early virologic response (undetectable HCV-RNA at week 12); pEVR = partial early virologic response (detectable but ≥2 $\log_{10}$ decrease in viral load at week 12); SVR = sustained virologic response.

Prognostic Value of Baseline FibroTest™ in the Diagnostic Population

- Baseline fibrosis stage estimated with FT had the same prognostic value as that estimated with biopsy for SVR and early virologic response (EVR).
- Five baseline factors were significantly associated with EVR (**Table 4**) and SVR (**Table 5**), in univariate and multivariate analyses: fibrosis stage estimated using FT or biopsy, genotype 2/3, baseline viral load, previous relapse, and previous treatment with non–PEG-IFN alfa.

Table 4. Prognostic Value of Baseline Factors for EVR in the Diagnostic Population

Factor	Univariate Analysis n = 1459: Odds Ratio (95% CI)	P	Multivariate Analysis[a] n = 1304: Odds Ratio (95% CI)	P
Fibrosis stage				
FibroTest™	4.9 (2.9-8.1)	<.0001	4.2 (2.2-7.9)	<.0001
Biopsy	1.2 (1.1-1.4)	.001	1.3 (1.1-1.5)	.001
Genotype 2/3	11.5 (7.9-16.6)	<.0001	8.9 (5.8-13.6)	<.0001
Baseline viral load <$\log_6$	1.9 (1.5-2.4)	<.0001	1.5 (1.2-2.0)	.003
Previous relapse (77 missing)	6.2 (4.8-8.0)	<.0001	6.5 (4.9-8.8)	<.0001
Previous non–PEG-IFN	1.3 (1.05-1.6)	.02	2.0 (1.5-2.7)	<.0001

CI = confidence interval; EVR = early virologic response; PEG-IFN = pegylated interferon.
[a] Patients with missing data for any of the 5 factors were excluded from the multivariate analysis.

Table 5. Prognostic Value of Baseline Factors for SVR

Factor	Univariate Analysis n = 678: Odds Ratio (95% CI)	P	Multivariate Analysis[a] n = 601: Odds Ratio (95% CI)	P
Fibrosis stage				
FibroTest™	4.5 (2.2-9.0)	<.0001	5.0 (2.3-11.0)	<.0001
Biopsy	1.5 (1.2-1.8)	<.0001	1.6 (1.3-2.0)	<.0001
Genotype 2/3	4.5 (3.1-6.6)	<.0001	4.0 (2.6-6.0)	<.0001
Baseline viral load <$\log_6$	1.7 (1.2-2.3)	.0009	1.6 (1.2-2.3)	.005
Previous relapse (77 missing)	1.6 (1.1-2.2)	.007	1.7 (1.1-2.4)	.007
Previous non–PEG-IFN	1.4 (0.99-1.9)	.06	1.6 (1.1-2.3)	.02

CI = confidence interval; PEG-IFN = pegylated interferon; SVR = sustained virologic response.
[a] Patients with missing data for any of the 5 factors were excluded from the multivariate analysis.

- Among patients with undetectable HCV-RNA at week 12 (n = 506), 2 factors were predictive of SVR by multivariate analysis: fibrosis estimated with FT or by biopsy and genotype 2/3 infection (**Table 6**).

Table 6. Prognostic Value of Baseline Factors for SVR Among Patients With Undetectable HCV-RNA at TW12 in the Diagnostic Population

Factor	Univariate Analysis n = 506: Odds Ratio (95% CI)	P	Multivariate Analysis[a] n = 444: Odds Ratio (95% CI)	P
Fibrosis stage				
FibroTest™	3.5 (1.5-8.0)	.003	4.0 (1.6-10.0)	.003
Biopsy	1.4 (1.1-1.7)	.003	1.5 (1.2-1.9)	.0007
Genotype 2/3	3.0 (2.0-4.6)	<.0001	2.9 (1.8-4.5)	<.0001
Baseline viral load <$\log_6$	1.7 (1.2-2.4)	.004	1.6 (1.08-2.4)	.02
Previous relapse (62 missing)	0.99 (0.7-1.4)	.96	1.06 (0.7-1.6)	.80
Previous non–PEG-IFN	1.3 (0.93-1.9)	.11	1.3 (0.8-2.0)	.23

CI = confidence interval; PEG-IFN = pegylated interferon; SVR = sustained virologic response; TW12 = treatment week 12.
[a] Patients with missing data for any of the 5 factors were excluded from the multivariate analysis.

- Incremental decreases in SVR were observed as METAVIR fibrosis scores increased when both FT and biopsy were used among all patients and among patients with undetectable HCV-RNA at TW12 (**Figure 2**).
- This trend was consistent across patients with METAVIR scores F0 and F1 by FT but not by biopsy.

Figure 2. SVR according to baseline fibrosis stage estimated using either FibroTest™ or biopsy. S = Armitage S for trend; SVR = sustained virologic response.

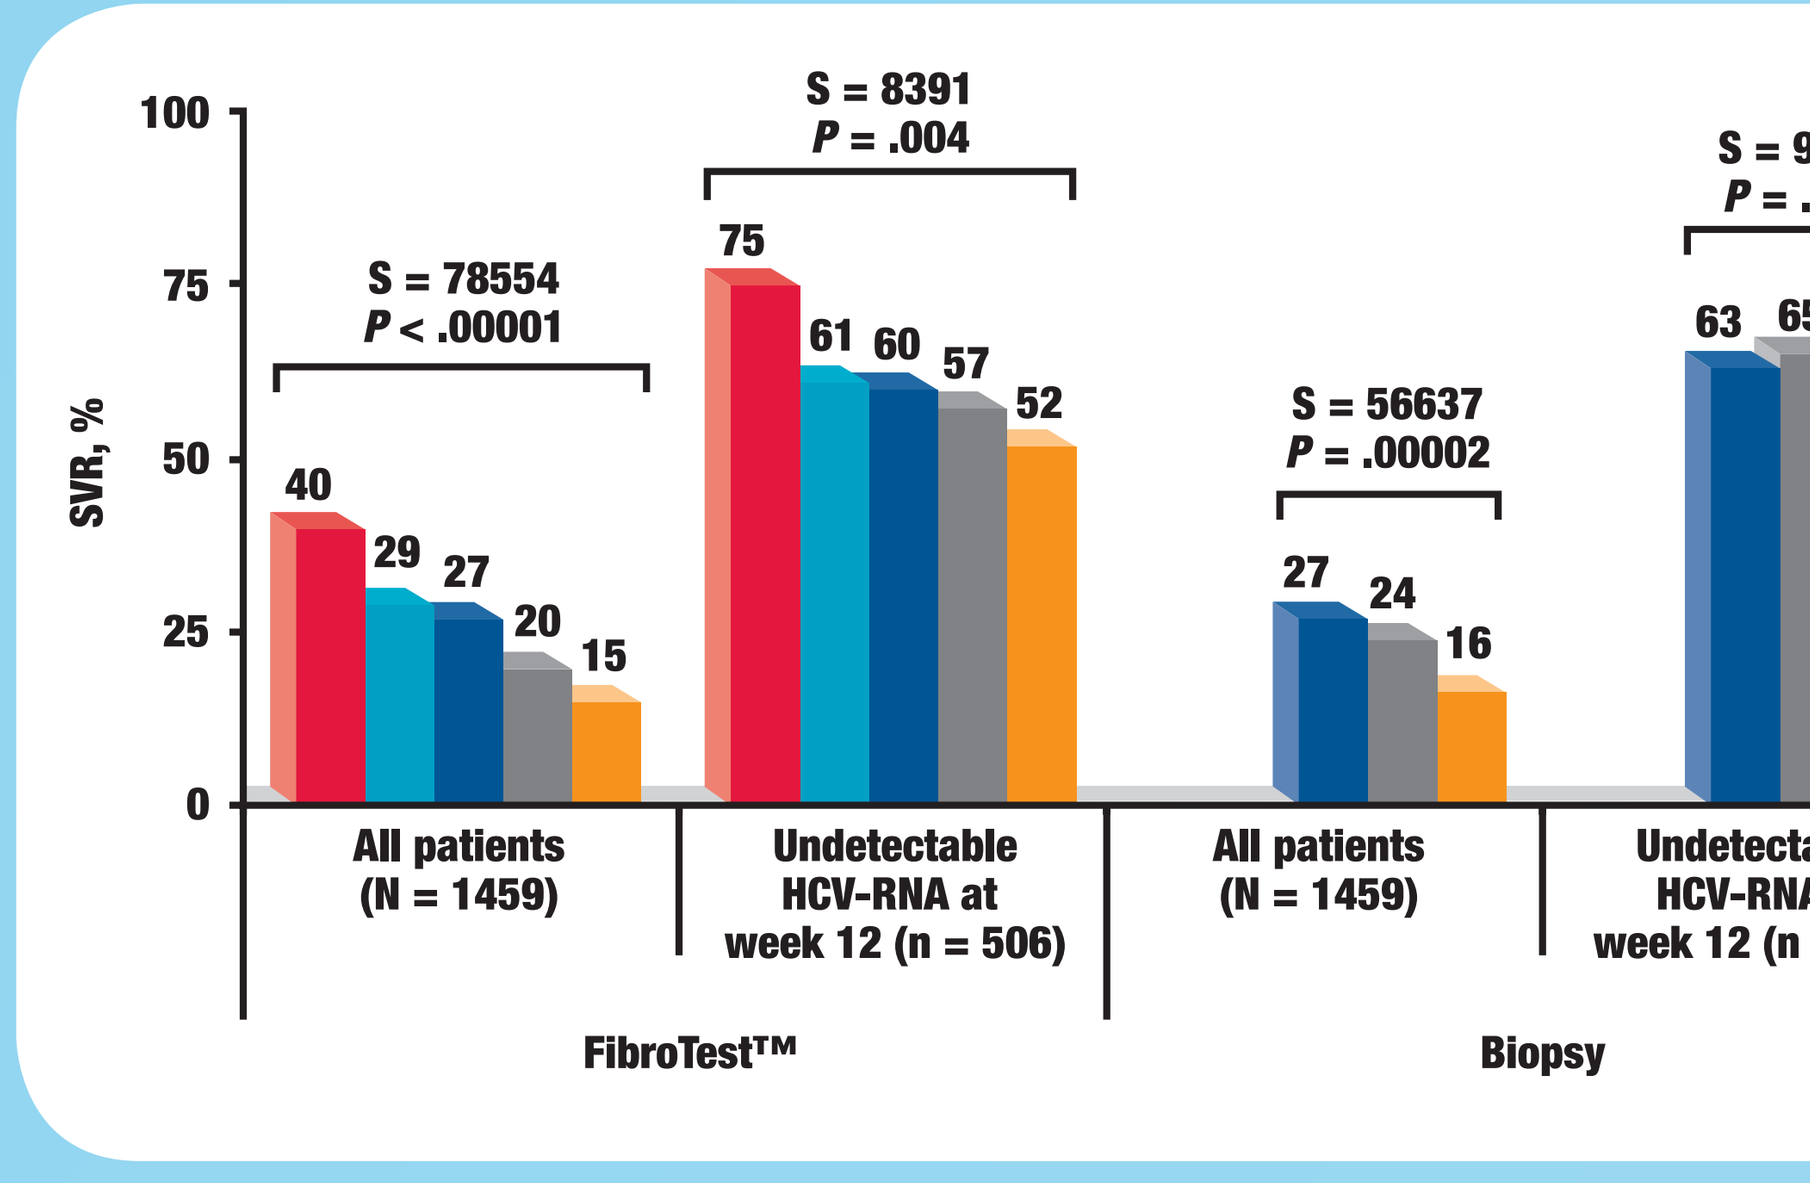
SVR, %
100
75
50
25
0
S = 78554
P < .00001
40
29
27
20
15
S = 8391
P = .004
75
61
60
57
52
S = 56637
P = .00002
27
24
16
S = 9224
P = .001
63
65
48
All patients
(N = 1459)
Undetectable
HCV-RNA at
week 12 (n = 506)
All patients
(N = 1459)
Undetectable
HCV-RNA at
week 12 (n = 506)
FibroTest™
Biopsy
F0
F1
F2
F3
F4

- Comparison of the differences between FT and biopsy results (**Figure 2**) suggests that a number of patients who had METAVIR score F2-F4 according to biopsy had METAVIR score F0-F1 liver disease according to FT.
- Patients whose fibrosis scores were overestimated by liver biopsy assessment were more likely to have attained SVR than predicted based on biopsy score.

Conclusions

- Results of this study suggest the diagnostic and prognostic utility of FT for clinicians as a simpler alternative to liver biopsy for patients who have chronic hepatitis C and in whom previous combination therapy failed.
- The prognostic value of FT for retreatment was similar to that reported in treatment-naive patients.
- The categorical nature of the biopsy scoring system renders weaker predictive power than does the dynamic FT system, which assesses a continuous variable and thus yields greater predictive power.
- This validated biomarker could be used as a surrogate marker for liver disease in trials evaluating the risk-benefit of maintenance therapy.

References

1. Manning DS et al. *Gastroenterology*. 2008;134:1670-1681.
2. Halfon P et al. *Gastroenterol Clin Biol*. 2008;32(suppl 1):22-39.
3. Ngo Y et al. *Clin Chem*. 2006;52:1887-1896.
4. Fontaine H et al. *Gastroenterol Clin Biol*. 2007;31:504-509.
5. Poynard T et al. *J Viral Hepatitis*. 2002;9:128-133.
6. Poynard T et al. *Hepatology*. 2003;38:481-492.
7. D'Arondel C et al. *J Viral Hepatitis*. 2006;13:182-189.
8. Patel K et al. *J Viral Hepatitis*. In press.
9. Poynard T et al. *Comp Hepatol*. 2004;3:8.
10. Poynard T et al. *Clin Chem*. 2007;53:1615-1622.
11. Poynard T et al. *Aliment Pharmacol Ther*. 2007;25:733-739.

Acknowledgments

We would like to thank Weiping Deng, Nicole Stauffer, Luminita Justice, Michele Sauer, Barbara Kapelan, Misti Linaberry, Kathleen Giordano, Stefanie Alfano, Peter Savino, Michael Salman, and Becky Liou of Schering-Plough Research Institute for their contributions to the conduct of the study.

Disclosures

TP, MC, ES, SF, WS, TB, EJH, AC, and MS are members of the Speakers Bureau for Schering-Plough; AC, EJH, and WS are members of the Speakers Bureau for Roche; TP, MC, ES, SF, TB, EJH, FG, MD, AC, and MS receive research support from Schering-Plough; AC receives research support from Roche; SF receives research support from Valeant and serves on the Speakers Bureau for Novartis; MC, JB, ES, EJH, and AC are consultants for Schering-Plough; AC is a consultant for Roche; PM, LG, MB, CB, and JKA are employees of SPRI and are stockholders of Schering-Plough; RT, RM-O, FC, TM, and PB have nothing to disclose. TP is the inventor of FibroTest™-ActiTest-SteatoTest and has a capital interest in BioPredictive, the company marketing the tests. The patents are held by Assistance Publique Hôpitaux de Paris, a public organization. MM is a full-time employee of BioPredictive.

Schering-Plough fully supported the study except for the FibroTest™ algorithm, which was supported by BioPredictive.

Presented at the 44th Annual Meeting of the European Association for the Study of the Liver, April 22–26, 2009, Copenhagen, Denmark